Enabling Integration of Alzheimer Disease (AD) Prevention Study Data by Application of CDISC Standards: Extension to Biometric Monitoring Device (BMD) Assessments

Stephen P. Arnerić [1], Volker D. Kern[1], Jon Neville[1], Jeffrey Kaye[2], Daniel Karlin[3], Jane Rhodes[4], Derek Hill[5], Ray Dorsey[6], Rebecca Spencer[7], Barrie Nelson[8], Michael Ibara[8], Jane Mohler[9], Lee Ryan[9], Jenny Barnett[10] and Lynn Hudson[1]

[1]Critical Path Institute; [2]Oregon Health & Science University; [3]Pfizer; [4]Biogen; [5]IXICO; [6]University of Rochester; [7]University of Massachusetts; [8]Clinical Data Interchange Consortium(CDISC); [9]University of Arizona; [10]Cambridge Cognition

Background and Objectives

In December 2016, the U.S. Food and Drug Administration (FDA) initiated the requirement of submitting data conforming to Clinical Data Interchange Standards Consortium (CDISC) standards for all new drug applications, indicating that clinical trials initiated at the present time must adopt these standards. CDISC standards facilitate aggregation of clinical data from diverse sources, which has enabled the FDA to improve the efficiency of integrated data analyses for new drug applications by ~40%. Biometric Monitoring Devices (BMDs) (e.g., wearables, smartphones, remote monitoring biosensors) offer the potential of measuring biologic events that may predict disease progression, or monitor key biologic functions in response to treatment interventions. The Coalition Against Major Diseases (CAMD), a nonprofit public-private-partnership within the Critical Path Institute (C-Path) focused on delivering Drug Development Tools that accelerate innovative treatments for Alzheimer disease (AD), is coordinating the development of these standards for integration into registration trials. BMDs may lead to clinical assessments sufficiently sensitive to detect changes in the pre-symptomatic stages of the neurodegenerative diseases like AD.

Methods

A partnership/collaboration between C-Path and the Arizona Alzheimer's Consortium was formed in 2016 to lay the foundation for additional data standardization and integration of data from prevention trials. On March 10, 2017, an international workshop was held to:

1. Evaluate the existing standards that apply to mobile devices that could be implemented in clinical drug trials and longitudinal disease progression studies;
2. Identify/prioritize existing gaps;
3. Develop a plan to accelerate the creation/implementation of CDISC standards required for future registration studies that assess three concepts-of-interest: mobility/ frailty, sleep, and cognitive performance.

Presentations and detailed notes from this meeting are available (LINK on the right).

Results

- Many foundational standards already exist thanks to the work of CDISC. CDISC launched the Shared Health And Research Electronic (SHARE) library for developing, integrating and accessing CDISC standards metadata electronically (Figure 1). SHARE is a curated resource that makes it easier to implement CDISC standards in electronic systems such as clinical data management systems, mobile apps, and learning health systems.

Figure 1 CDISC SHARE library overview (Ref. 1)

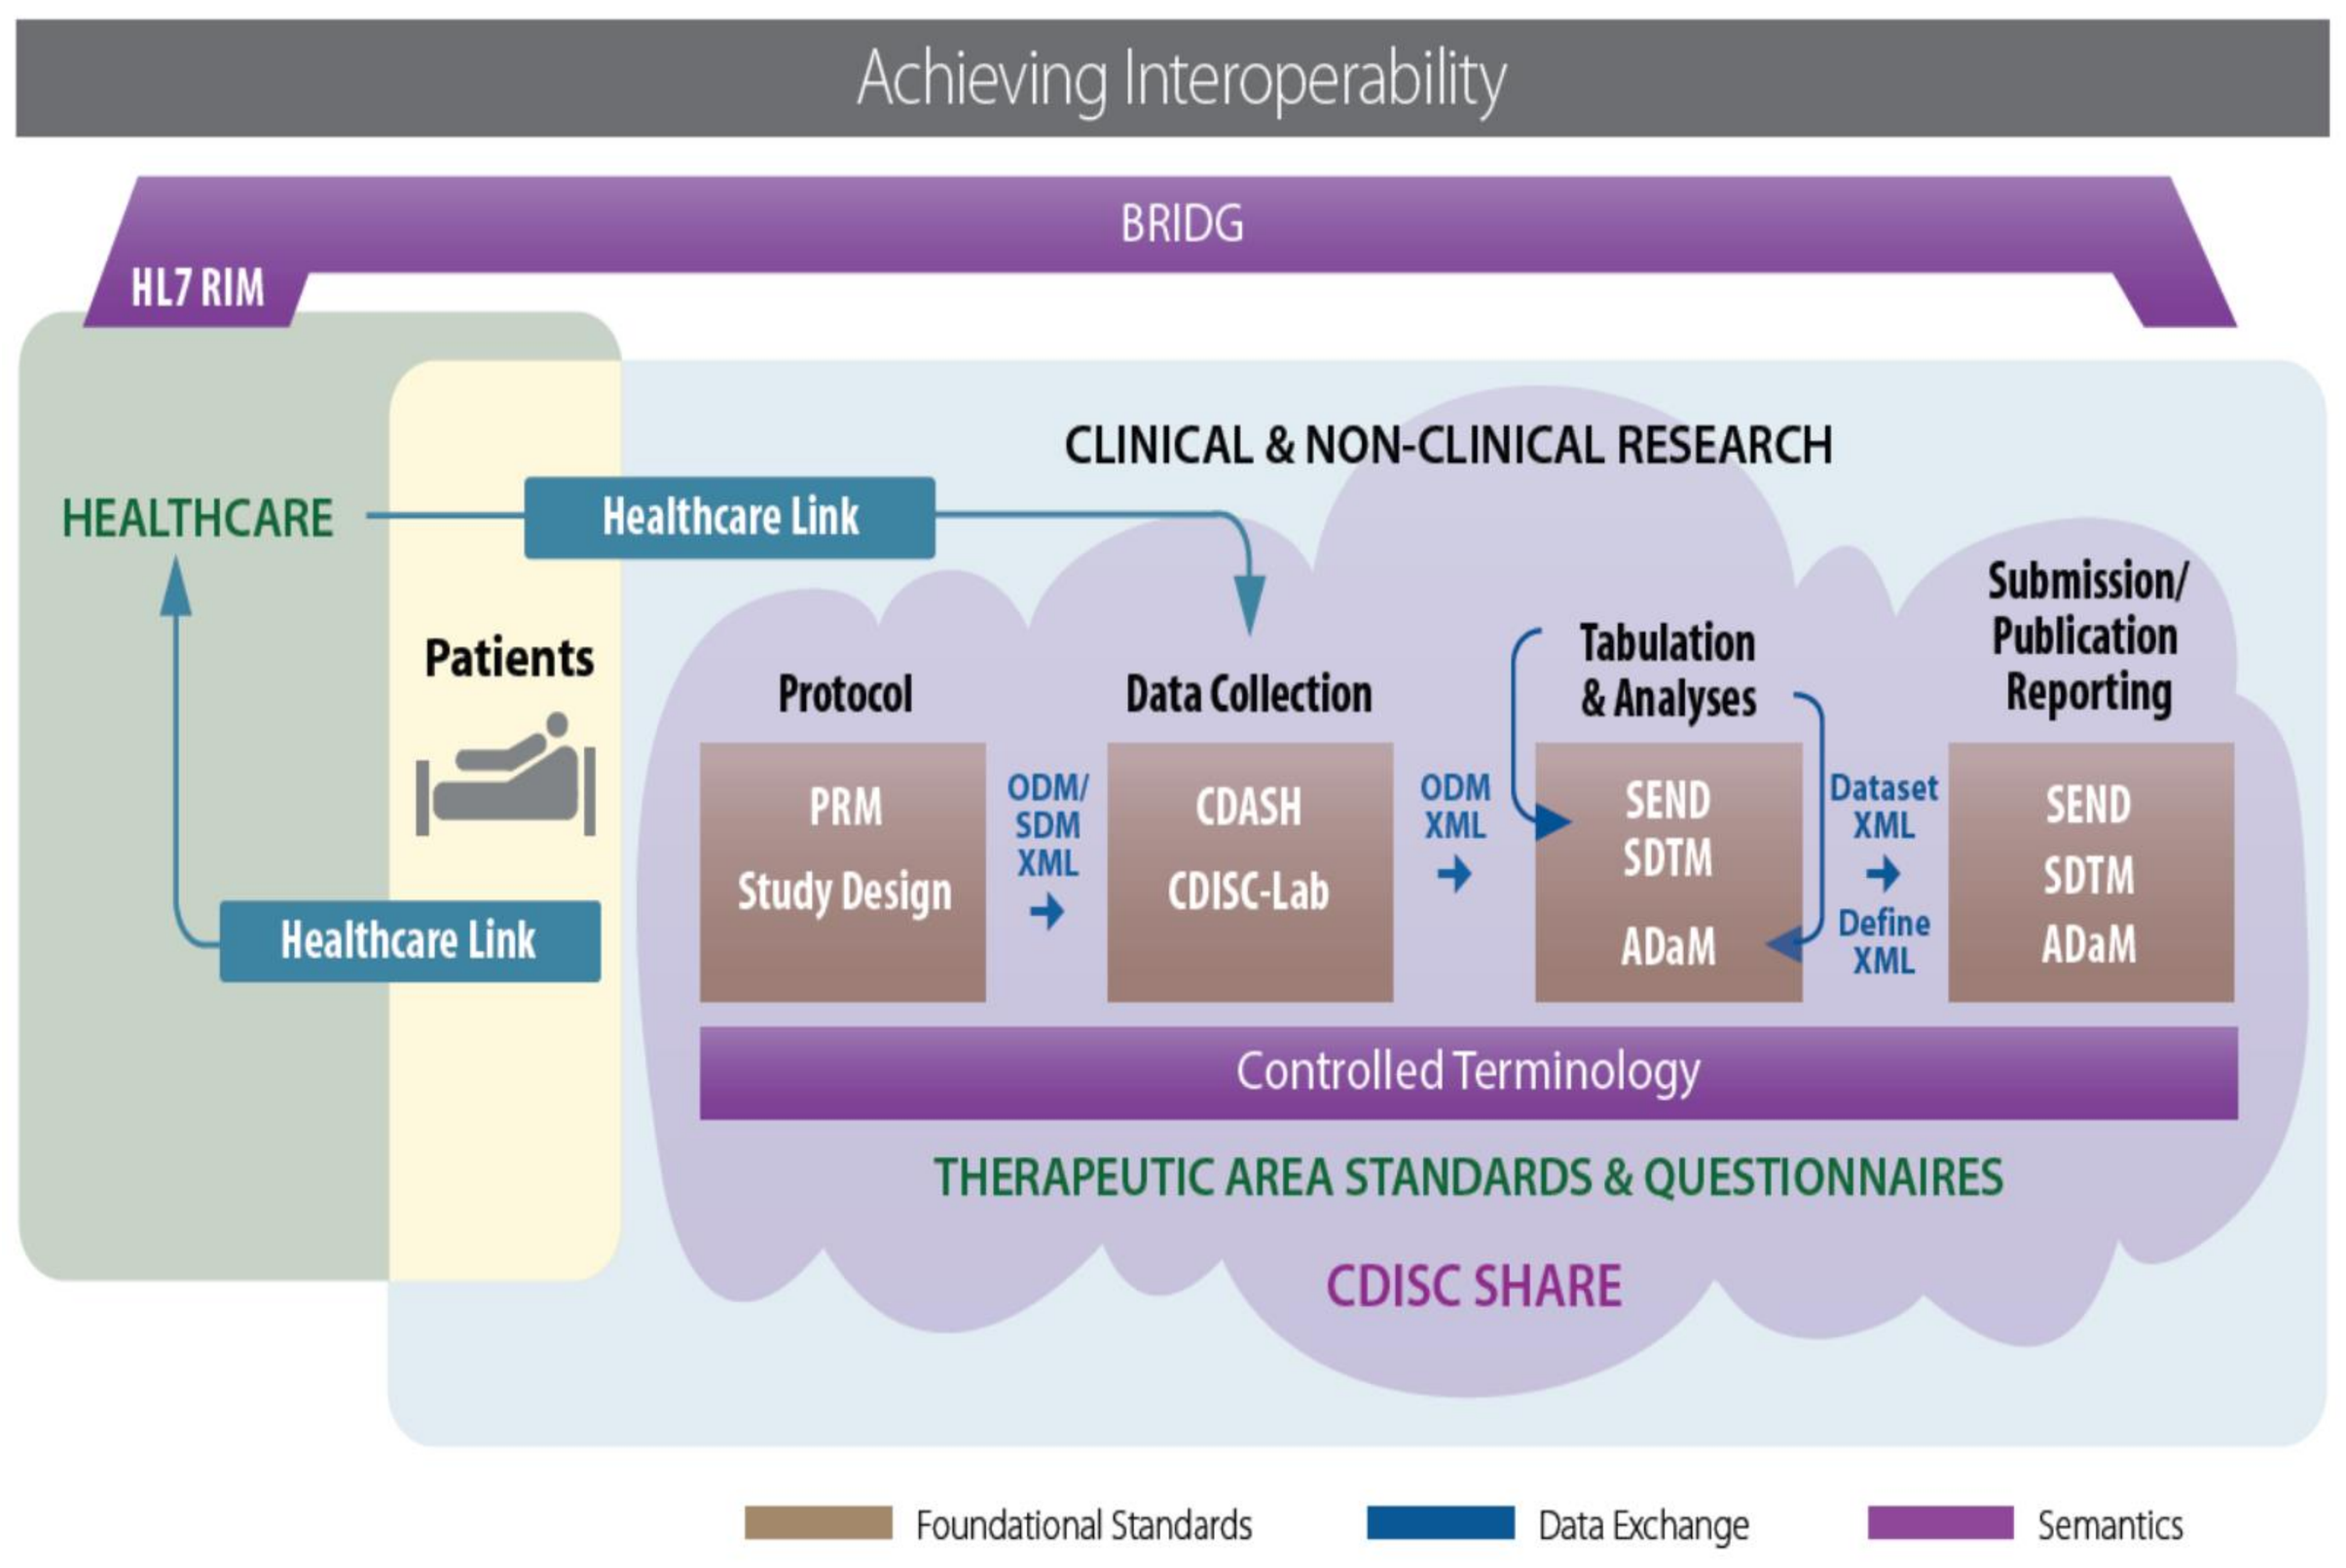
Achieving Interoperability
BRIDG
HL7 RIM
HEALTHCARE
Healthcare Link
Patients
Healthcare Link
CLINICAL & NON-CLINICAL RESEARCH
Protocol
PRM
Study Design
ODM/
SDM
XML
Data Collection
CDASH
CDISC-Lab
ODM
XML
Tabulation
& Analyses
SEND
SDTM
ADaM
Dataset
XML
Define
XML
Submission/
Publication
Reporting
SEND
SDTM
ADaM
Controlled Terminology
THERAPEUTIC AREA STANDARDS & QUESTIONNAIRES
CDISC SHARE
Foundational Standards
Data Exchange
Semantics

- Increasingly, clinical trials in neurodegenerative diseases are focusing on the early stages of the disease (Figure 2), trying to identify individuals who are genetically, or otherwise susceptible prior to developing any symptoms (Ref. 2 and 3).
- High-level considerations were developed for all three concepts-of-interest (COIs), and how they interrelate (Figure 3). Ideally these COIs and corresponding standards, could be developed across neurodegenerative diseases.
- More detailed considerations of data flow were outlined for each COI (Figure 4).
- Additional work needs to be completed in detailing data provenance, annotating contextual metadata, and compliance with Good Clinical Practices (GCP) to support the requirements for regulator submissions (Figure 5).

Figure 2
Alzheimer disease (AD) Stages: Framing the dilemma of what to measure and when

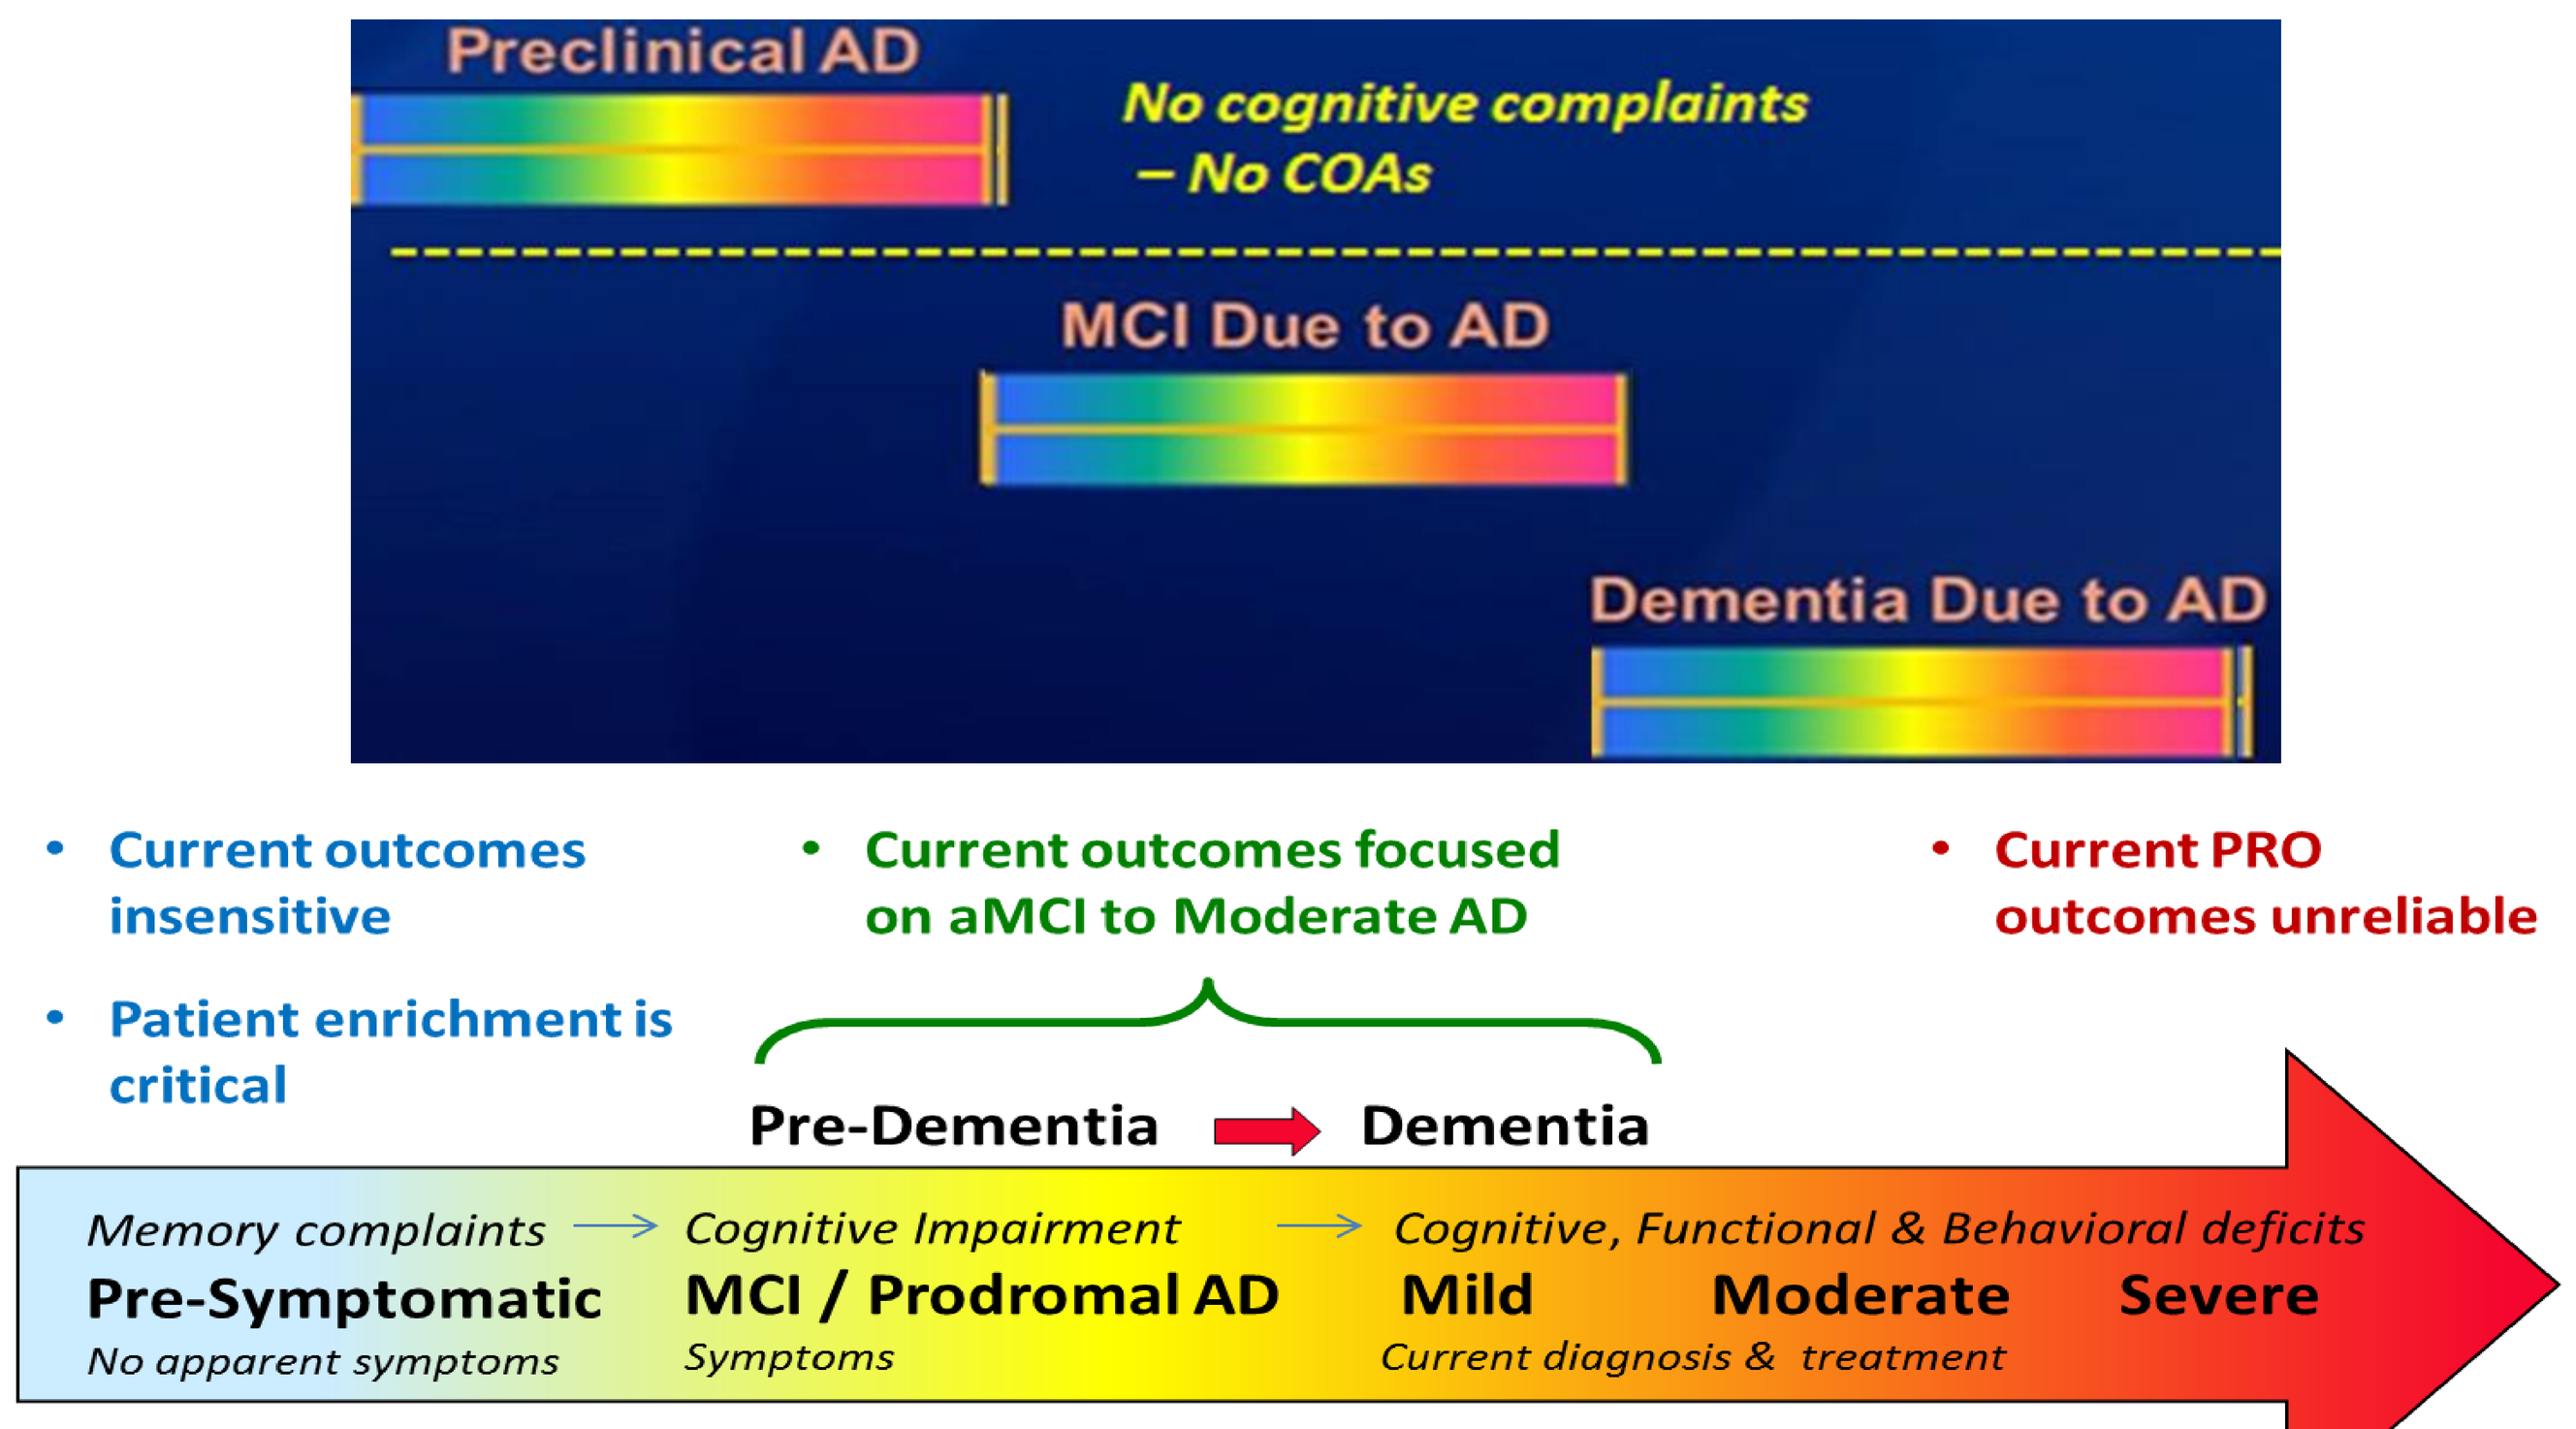
Preclinical AD
No cognitive complaints
– No COAs
MCI Due to AD
Dementia Due to AD
Current outcomes insensitive
Patient enrichment is critical
Current outcomes focused on aMCI to Moderate AD
Current PRO outcomes unreliable
Pre-Dementia
Dementia
Memory complaints
Pre-Symptomatic
No apparent symptoms
Cognitive Impairment
MCI / Prodromal AD
Symptoms
Cognitive, Functional & Behavioral deficits
Mild
Moderate
Severe
Current diagnosis & treatment

Figure 3
High level Concepts-of-Interest applicable across neurodegenerative diseases

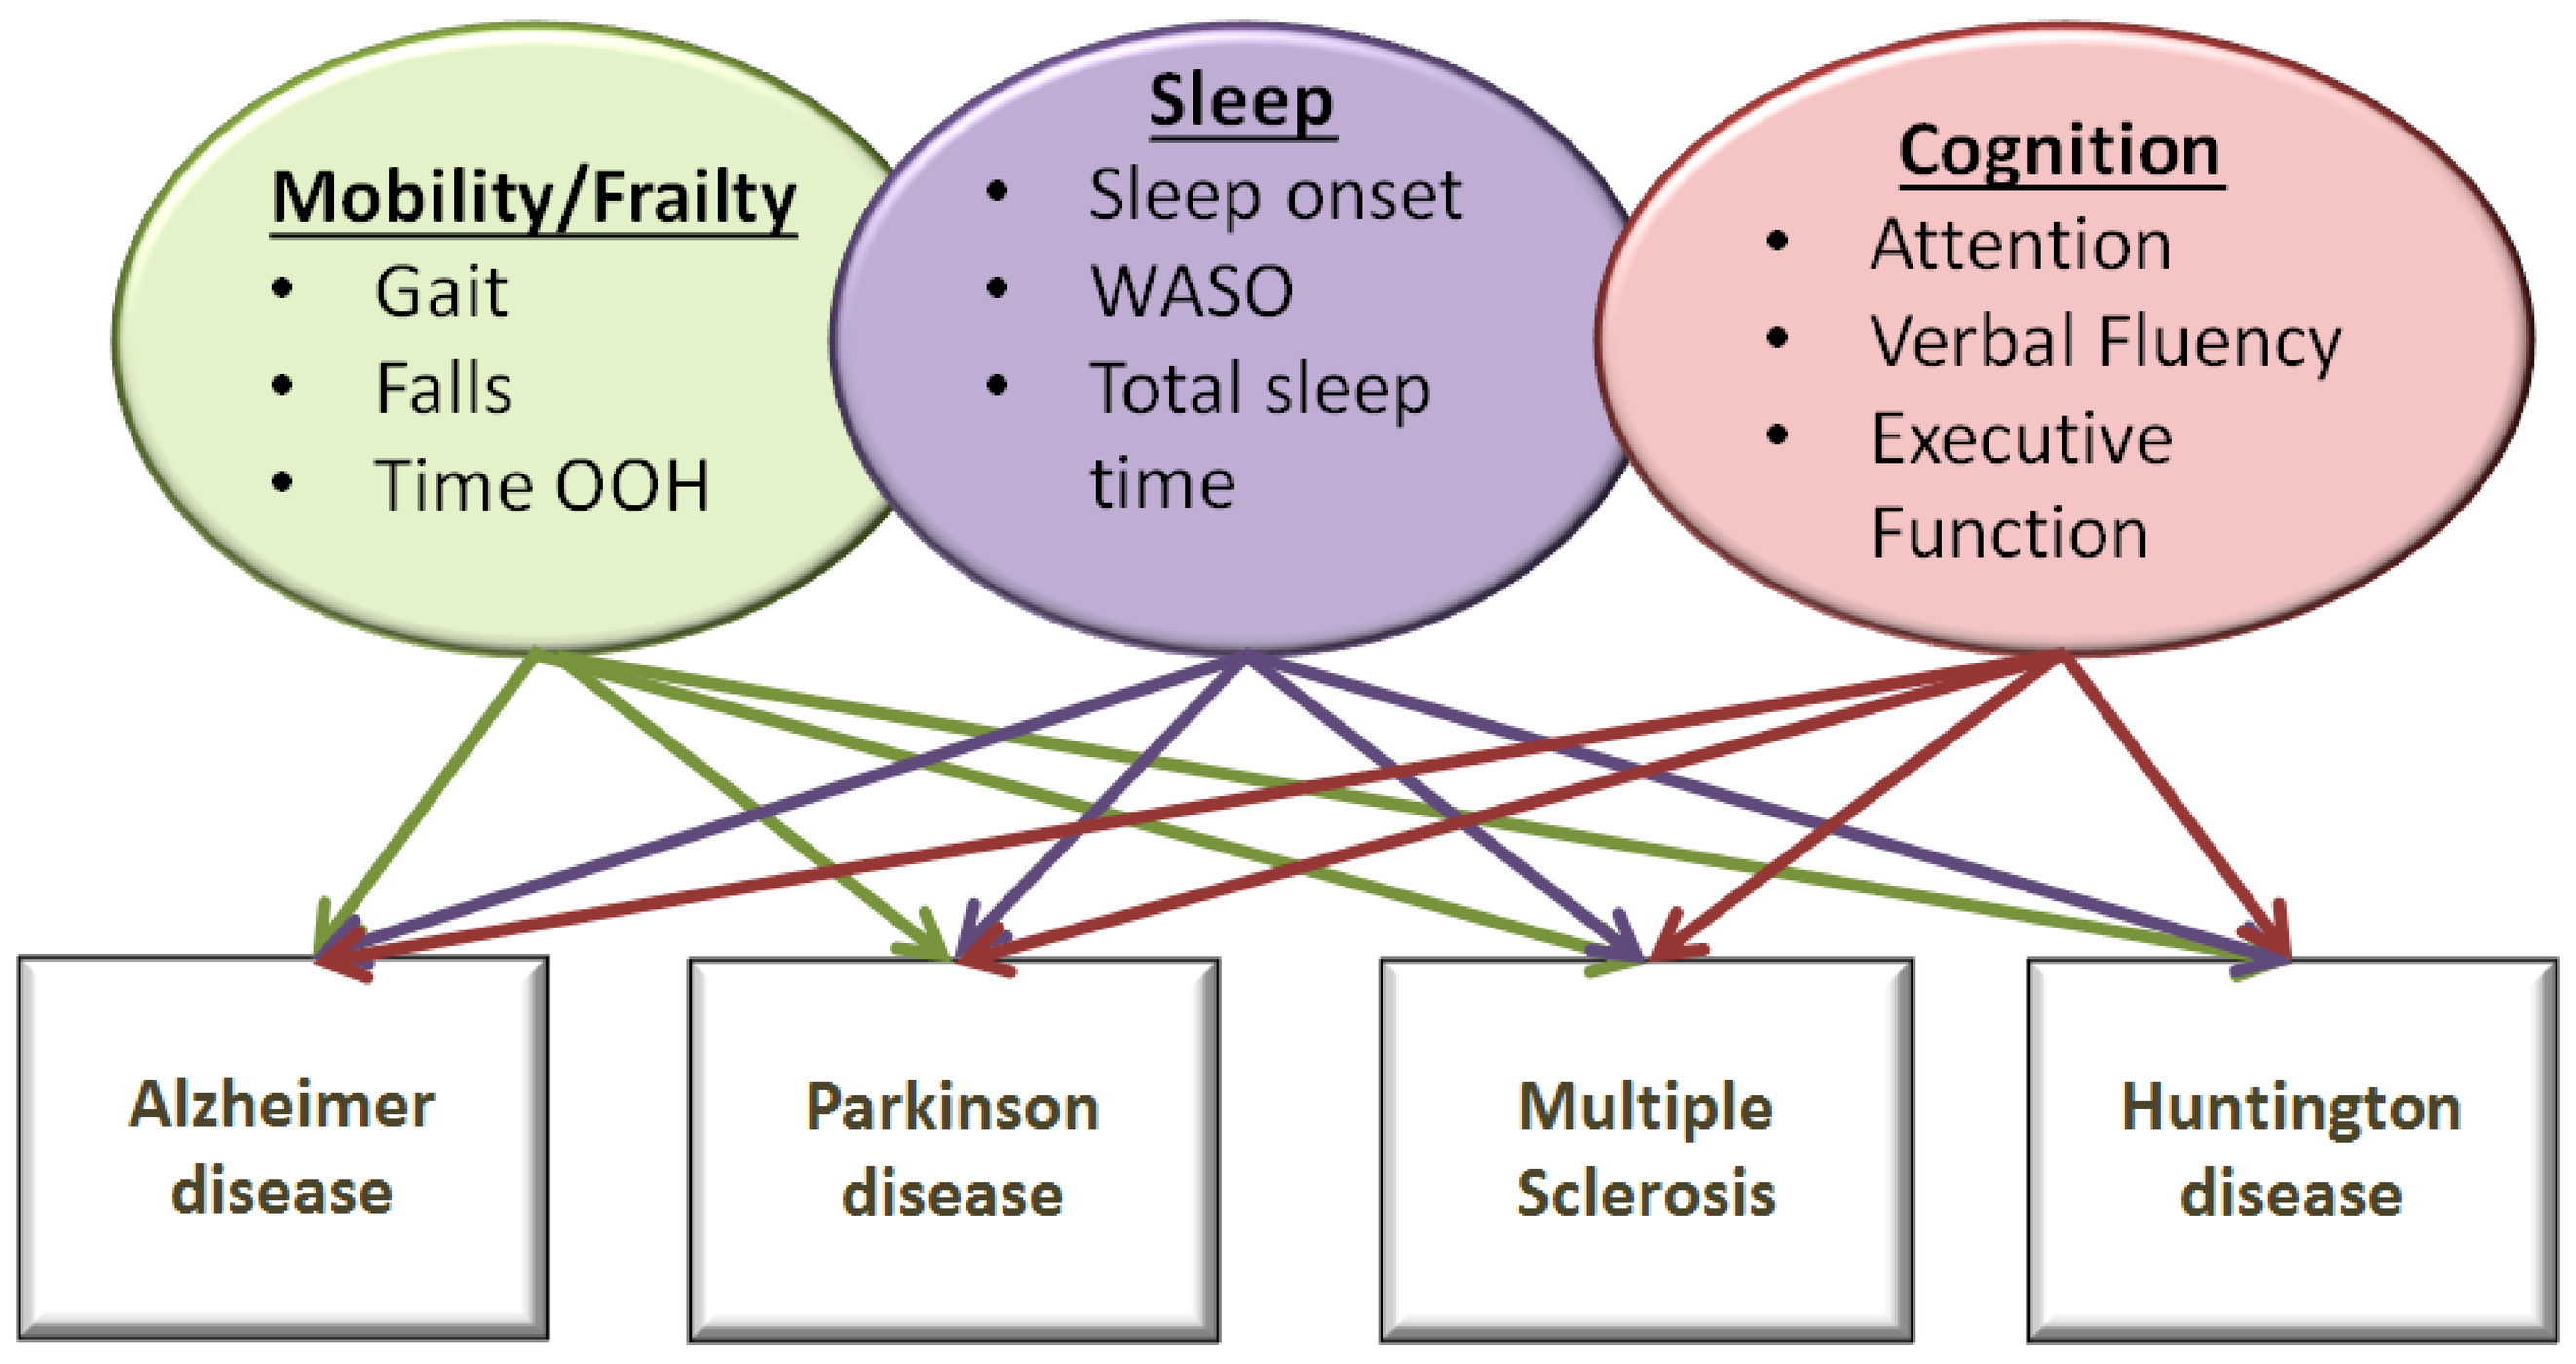
Mobility/Frailty
Gait
Falls
Time OOH
Sleep
Sleep onset
WASO
Total sleep time
Cognition
Attention
Verbal Fluency
Executive Function
Alzheimer disease
Parkinson disease
Multiple Sclerosis
Huntington disease

Figure 4
Data flow from Biometric Monitoring Devices (BMDs) assessing specific measures for each Concept-of-Interest: High-level information capture

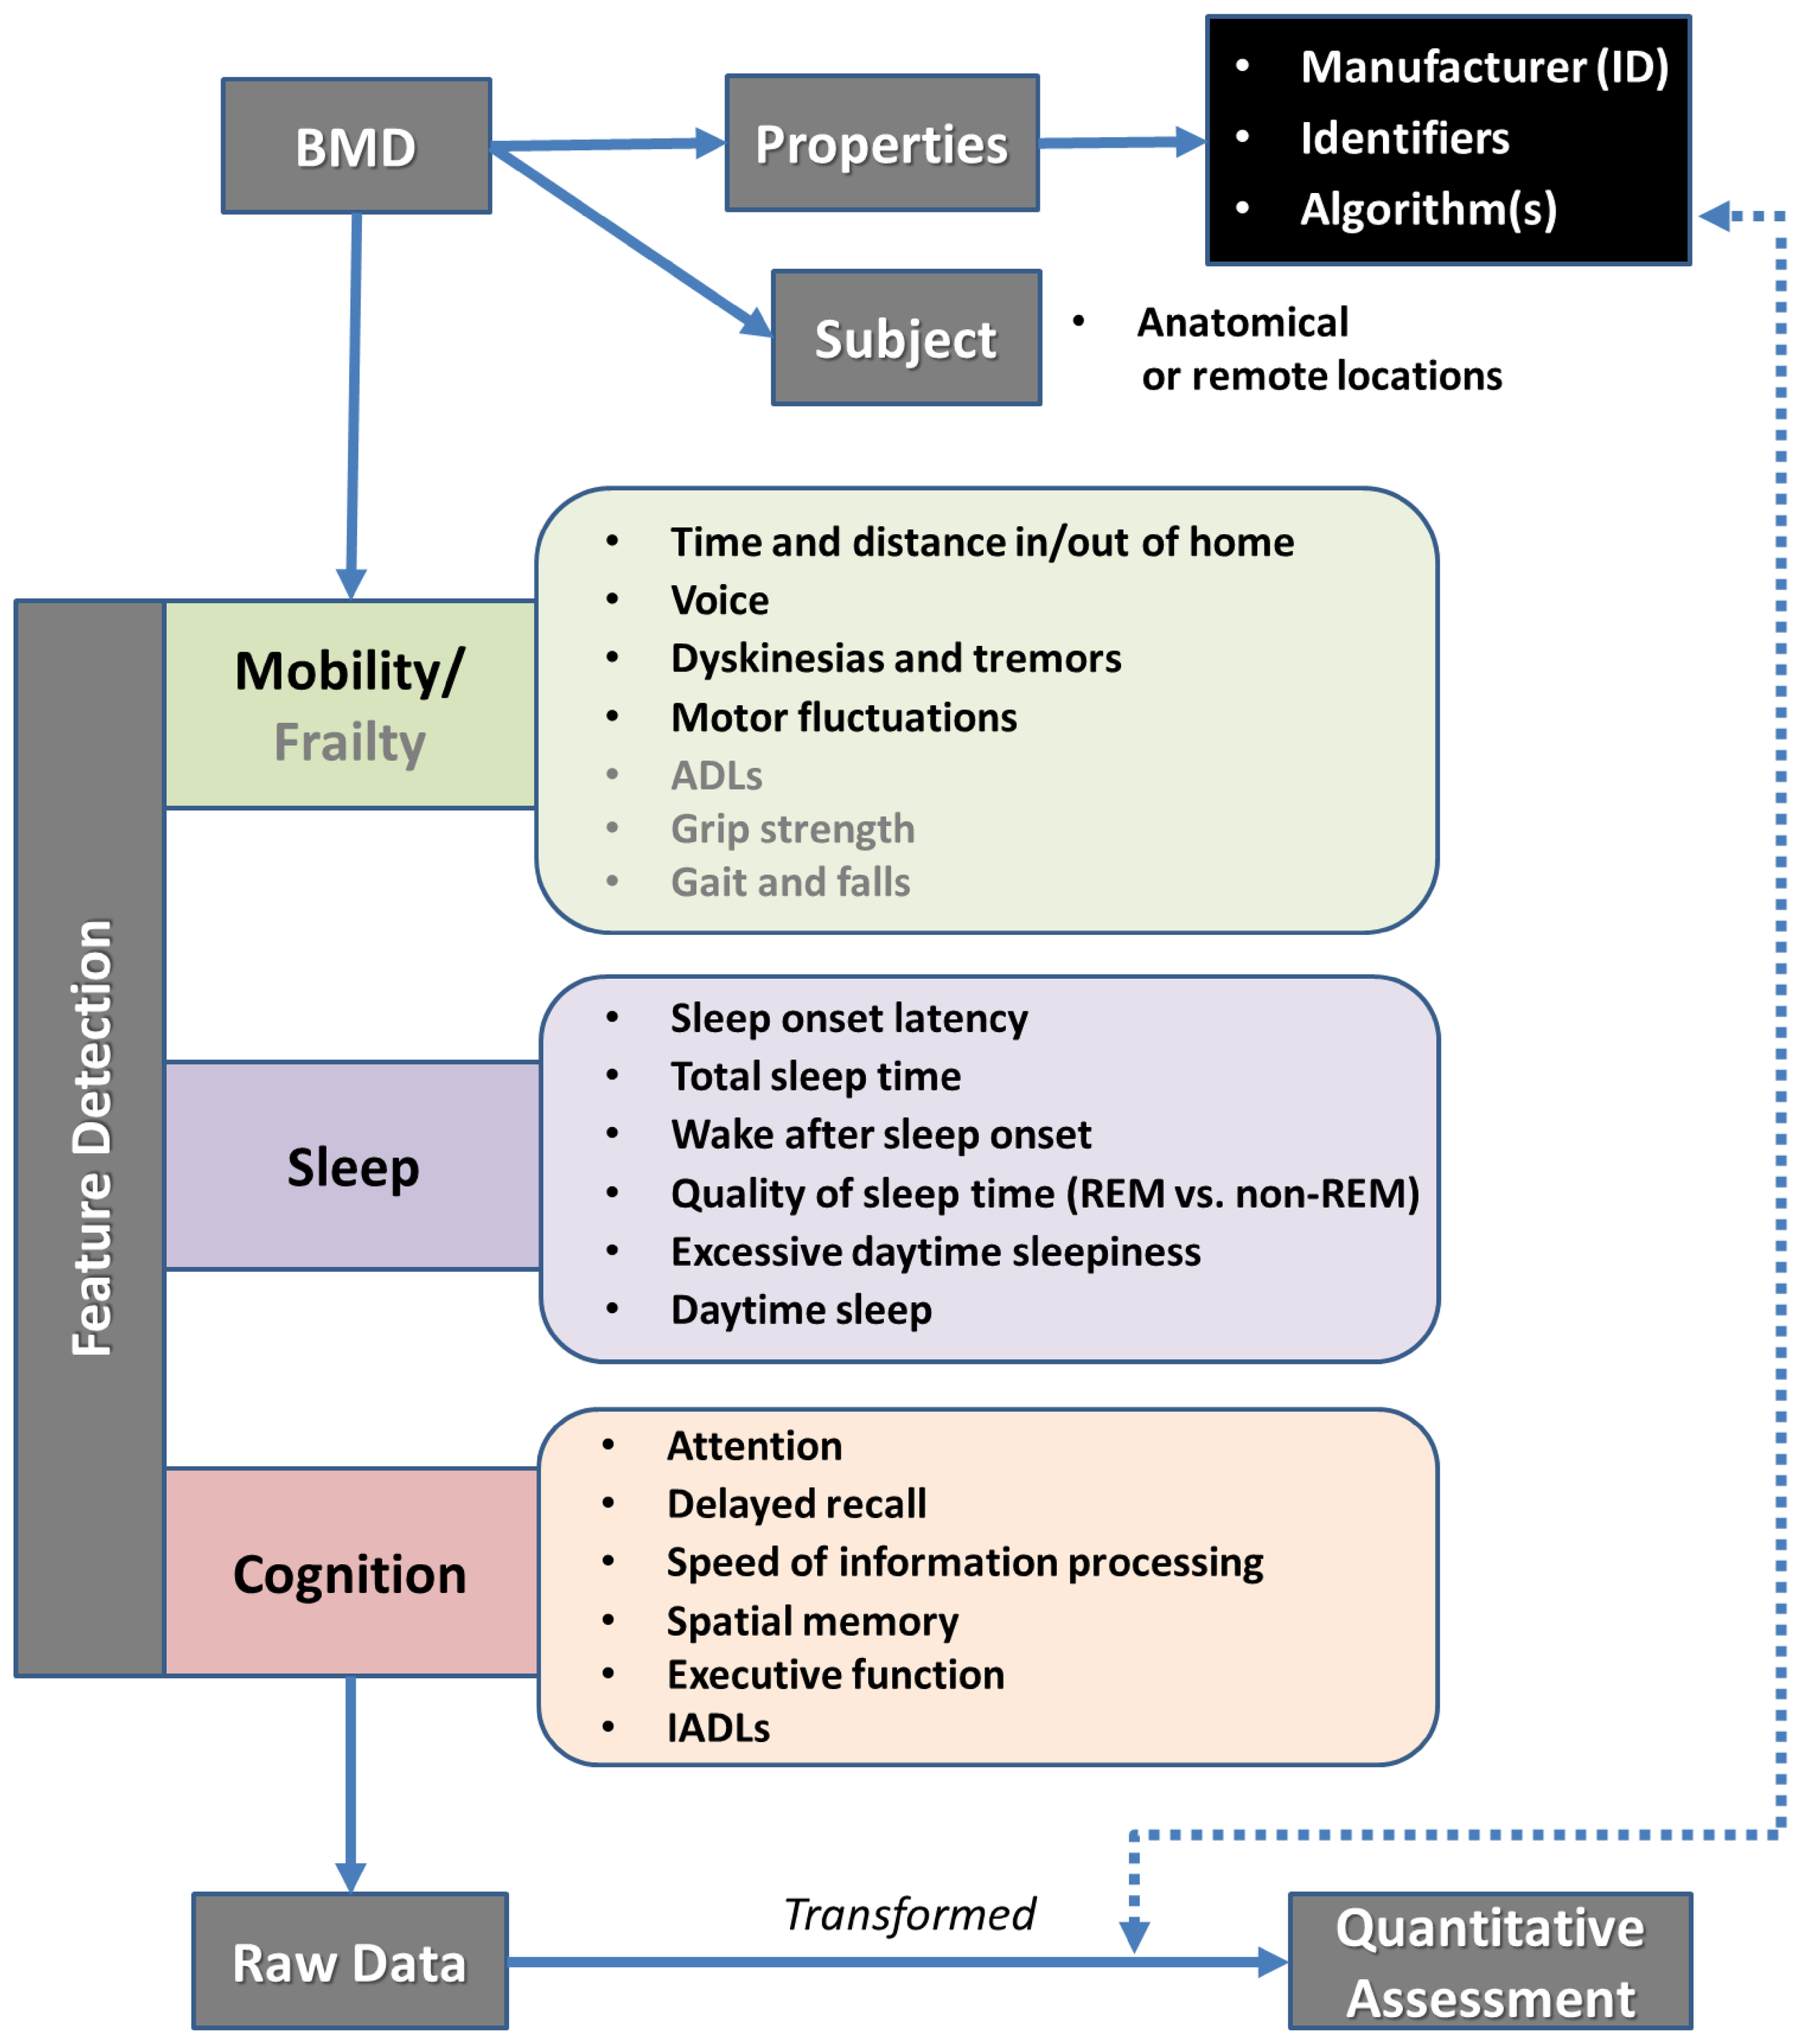
BMD
Properties
Manufacturer (ID)
Identifiers
Algorithm(s)
Subject
Anatomical or remote locations
Feature Detection
Mobility/ Frailty
Time and distance in/out of home
Voice
Dyskinesias and tremors
Motor fluctuations
ADLs
Grip strength
Gait and falls
Sleep
Sleep onset latency
Total sleep time
Wake after sleep onset
Quality of sleep time (REM vs. non-REM)
Excessive daytime sleepiness
Daytime sleep
Cognition
Attention
Delayed recall
Speed of information processing
Spatial memory
Executive function
IADLs
Raw Data
Transformed
Quantitative Assessment

Figure 5
Many Aspects of the infrastructure required to understand disease progression and treatment impact in clinical drug trials already exist. (from CDISC 2017 Training Materials)

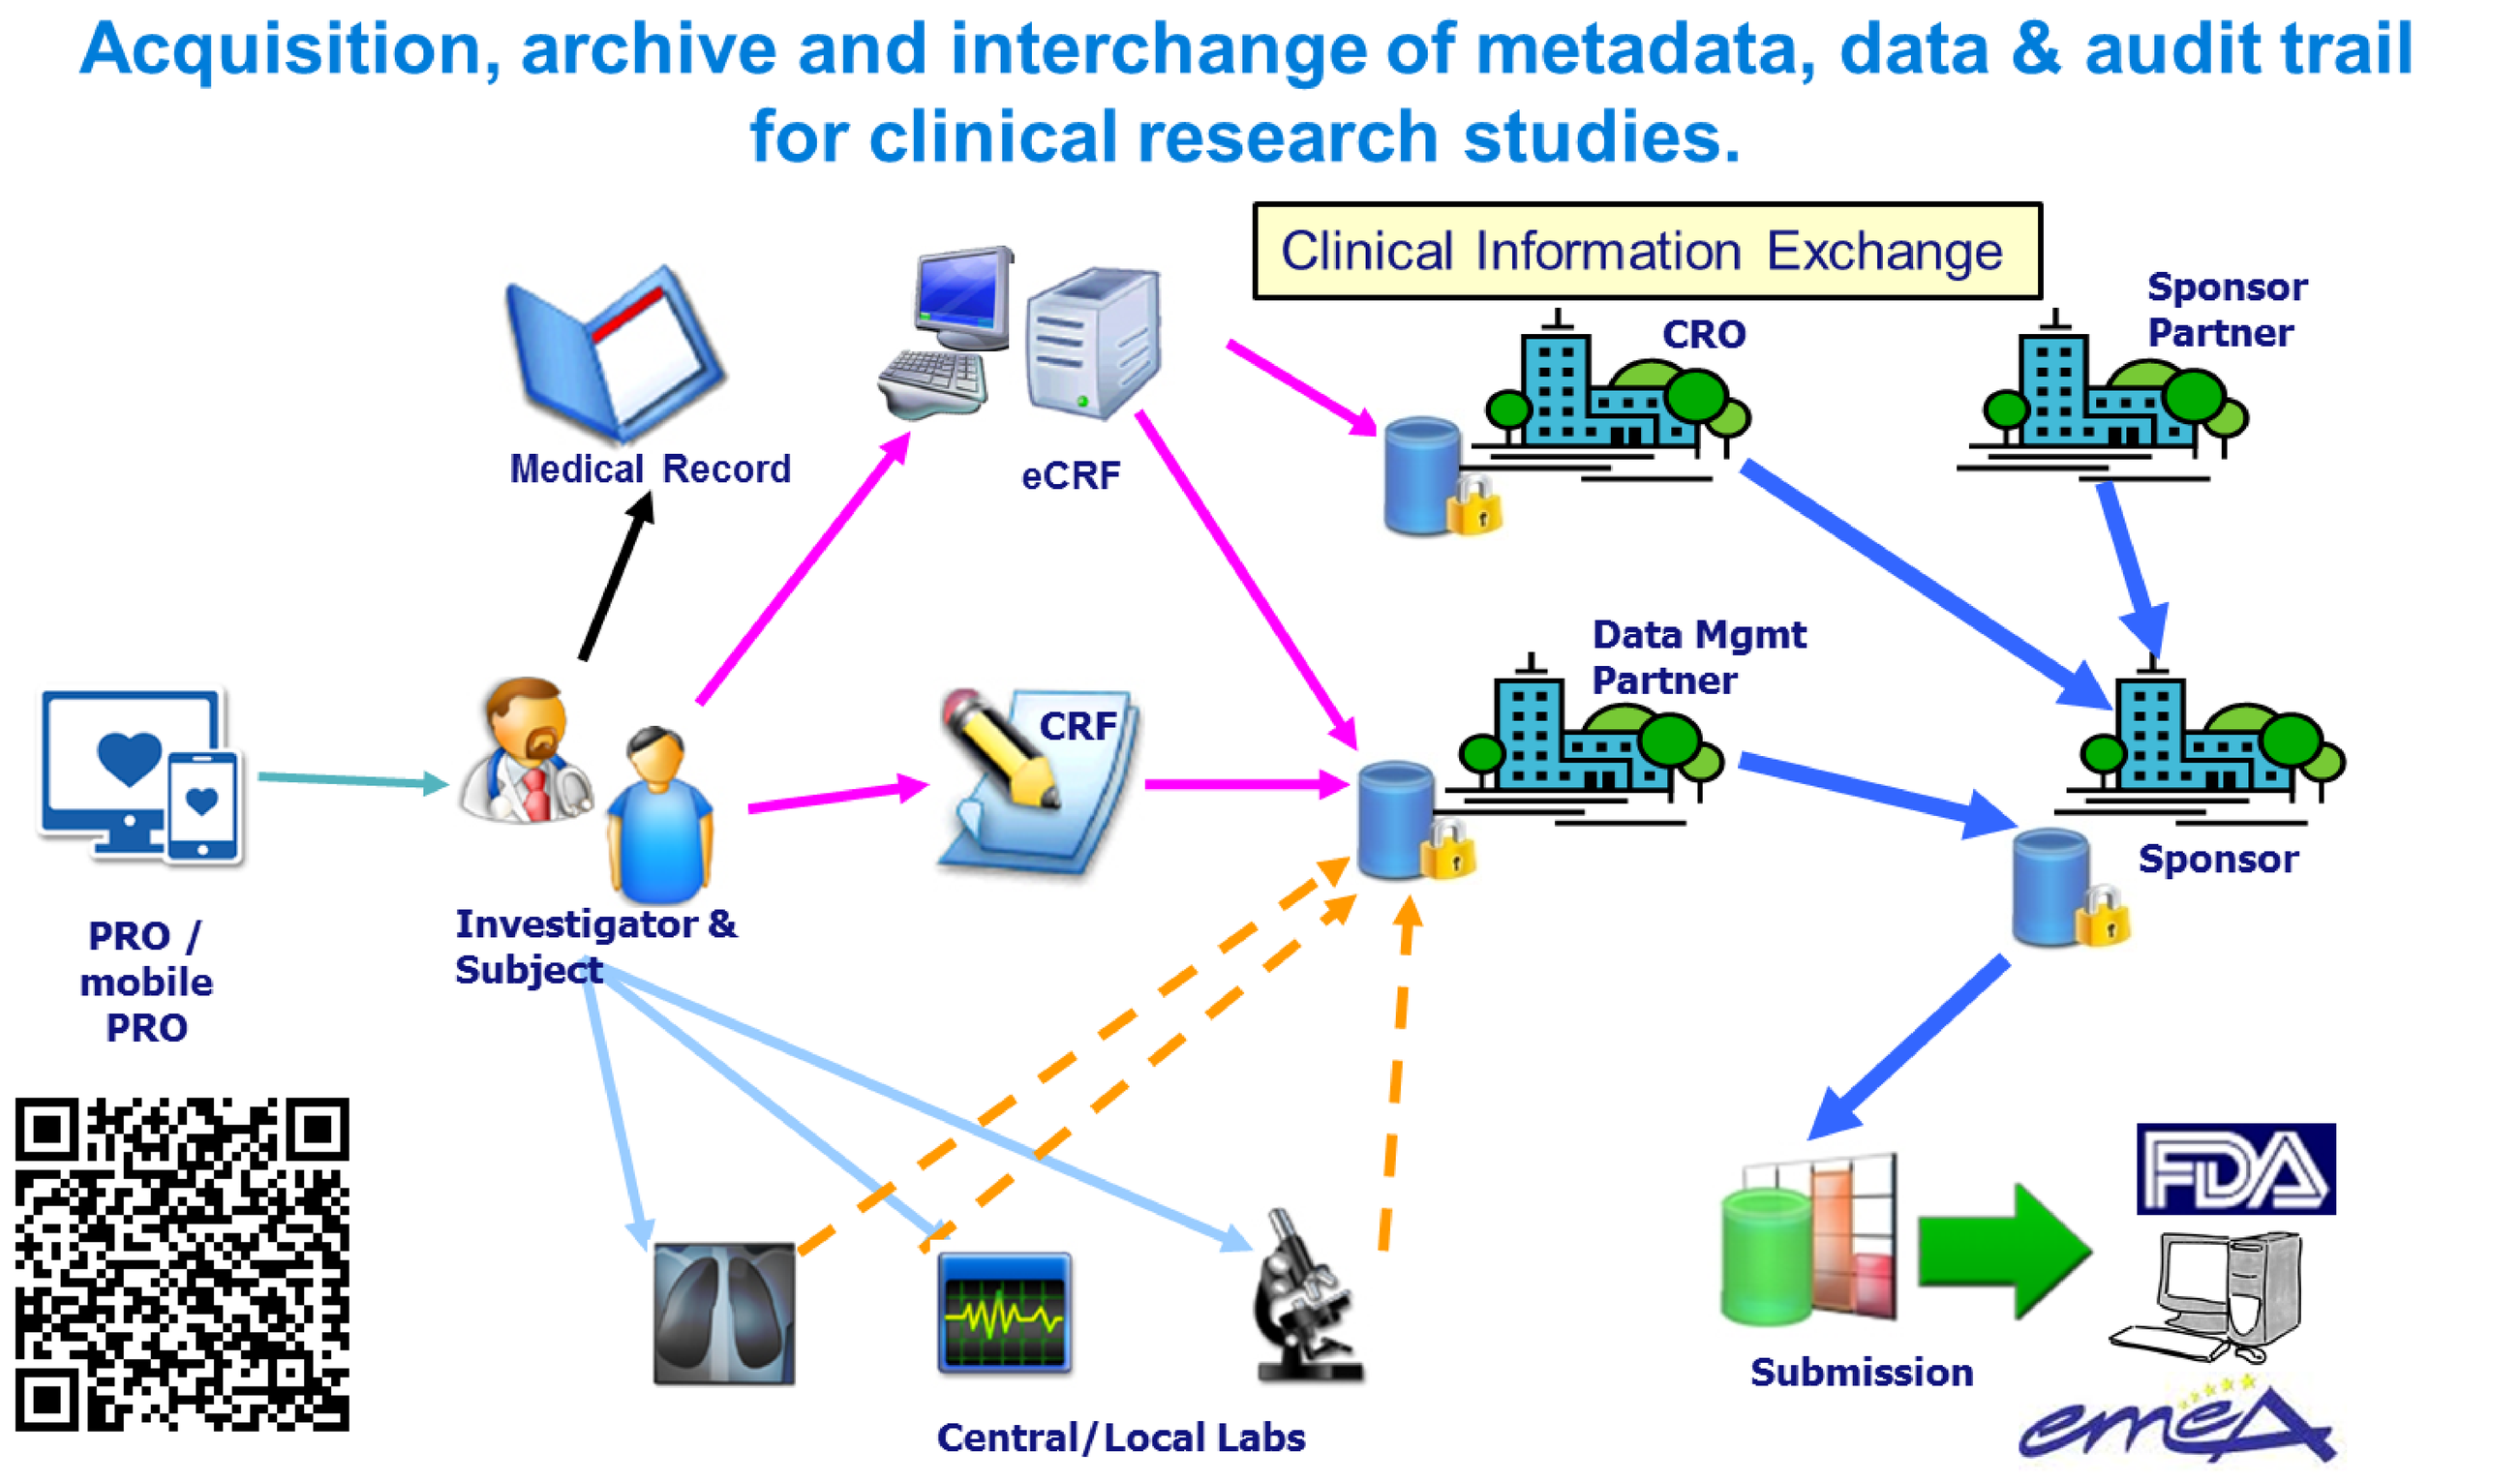
Acquisition, archive and interchange of metadata, data & audit trail for clinical research studies.
Clinical Information Exchange
Medical Record
eCRF
CRO
Sponsor Partner
Data Mgmt Partner
CRF
PRO / mobile PRO
Investigator & Subject
Sponsor
Central/Local Labs
Submission
FDA
emea

Discussion

- Continuous and passive monitoring of patient outcomes will likely provide greater ecological validity, improve statistical power to facilitate personalized therapeutics, reduce cost of lengthy trials, and enable tailored therapeutic approaches. This has great potential to facilitate clinical trial design, and reduce costs for preventative therapy trials started in the pre-symptomatic stages of AD.
- Many foundational data standards exist that will support the acquisition, tabulation, analysis and final reporting of data to the FDA (Figure 1). Some specific standards to align correlative metadata will likely be required.
- Understanding what dimensions of Quality-of-Life are most important to patients and their caregivers may help focus which biosensor assessment to focus on.
- Cross correlation of mobility/frailty, sleep, and cognition assessments may lead to more sensitive ways to understand disease progression and treatment responses.
- Various stakeholders will need to effectively collaborate to develop, validate, and secure regulatory endorsement of the use of Biometric Monitoring Devices in clinical trials for patients with AD and other neurologic disorders.

Conclusions

1. In the absence of more sensitive, regulatory-qualified patient assessment technologies, large, lengthy prevention trials (5-10 years) will be required to progress to MCI stages where existing assessments are available (Figure 2).
2. Developing and validating specific Use-Cases are required to gain confidence in their utility as viable Drug Development Tools.
3. Development of data repositories of structured data, using CDISC standards gathered from well-characterized patient populations, will be foundational to supporting the well defined Contexts-of-Use.
4. Key Gaps Remaining:
 - Capture critical contextual metadata to enable interpretation;
 - Use of uniform terminology regarding sensor, device, and measurement should be a focus;
 - High priority should be given to understanding which functional outcomes patients and caregivers prefer, and how these Concepts-of-Interest might relate to features captured by BMDs (those interested in contributing to CAMD's survey ***"The Voice of Those Who Care"***, please follow link on the right);
 - While the payer's perspective should not be ignored, this topic could be a later priority when alignment with regulators has been achieved.

References

(1) Becnel, L. (2017) S28: An overview of CDISC standards and services, American Medical Informatics Association, 3/29/17, San Francisco.
(2) Gray, B.B. (2016) Signs Before Symptoms, *NeurologyNow*, June/July, pp 33-36.
(3) Mortamais, M. *et al.* (2017) Detecting cognitive changes in preclinical Alzheimer's disease: A review of its feasibility, *Alzheimer's & Dementia*, 13:468-492.

Acknowledgments: This work was supported by a grant the Arizona Alzheimer's Consortium and, in part, by grant number 1U18FD005320 from the U.S. Food and Drug Administration's Critical Path Public Private Partnerships Grant, and members including: AbbVie Inc., Alzheimer's Association, Alzheimer's Disease Discovery Foundation, Alzheimer's Research UK, Biogen, Boehringer Ingelheim, CHDI Foundation, Eisai, Eli Lilly, F. Hoffmann La Roche, Merck Sharp & Dohme, Novartis Pharmaceuticals Corporation, Pfizer Inc., Takeda Pharmaceuticals, and UsAgainstAlzheimer's.